Development of mucoadhesive drug delivery systems by precipitation of chitosan on drug-loaded microparticles

D. Preisig[1], M. Weingartner[1], F. Varum[2], R. Bravo[2], R. Alles, J. Huwyler[1], M. Puchkov[1]

[1] *Department of Pharmaceutical Sciences, University of Basel, Basel, Switzerland*
[2] *Tillotts Pharma AG, Rheinfelden, Switzerland*

INTRODUCTION

Multiparticulate dosage forms consist of small sub-units containing the drug and having the advantage of more uniform and predictable transit time, better distribution and less local irritation in the gastrointestinal tract, compared to single unit dosage forms [1, 2]. These multiparticulates, when designed as mucoadhesive formulations, can potentially prolong and normalize the residence time in the gastrointestinal tract, and increase oral drug bioavailability due to their adhesive interactions with the gastrointestinal mucus [3]. Examples of methods for preparation of mucoadhesive pellets and microspheres are extrusion–spheronization [1], spray-drying [4], and emulsion-solvent evaporation [5].

Here, we introduce the use of a novel excipient based on calcium carbonate (FCC, Omyapharm) as a drug carrier and adsorption platform of chitosan. FCC has a small particle size (5-15 µm) and a high porosity (70%, v/v), and can be loaded with various drugs by solvent evaporation [6]. Among other polymers, chitosan is well known for its good mucoadhesive properties [7]. Precipitation of chitosan on calcium carbonate templates for the preparation of hollow microcapsules was recently described [8], but not intended to be used as mucoadhesive multiparticulates in order to increase residence time in the human gastrointestinal tract. Furthermore, the dissolution of the calcium carbonate core is not intended in the approach here presented.

AIMS

- Preparation of **mucoadhesive microparticles,** based on drug-loaded FCC using a chitosan precipitation method for development of a multiparticulate drug delivery system.
- Evaluation of in vitro mucoadhesion using a **particle-retention assay**, simulating the conditions in the human colon. For this purpose, a new method for precise quantification of detached calcium carbonate microparticles was developed.

CONCLUSIONS

- The precipitation method presented here is suitable for the preparation of chitosan-coated microparticles.
- Increasing the chitosan content in the microparticles resulted in increased particle retention *in vitro*.
- Drug release quantification from the flow-channel experiments closely correlated with data obtained from USP IV experiments.

MATERIALS & METHODS

Particle preparation

Drug loading: Metronidazole benzoate (MBZ) was used as model drug for the preparation of mucoadhesive drug loaded microparticles. Drug loading into FCC was performed by solvent evaporation according to Preisig *et al*. [6]. Theoretical drug load was 40% (w/w).

Precipitation method: The MBZ-loaded FCC particles were used for mucoadhesive coating by precipitation of chitosan (Figure 2). The chitosan solutions were prepared in diluted acetic acid by stirring for 12 h and adjusting to pH 5. The MBZ-loaded FCC particles (10.0 g) were dispersed in 1 L of chitosan solution, and NaOH (0.05M) was slowly added (0.5 mL/min) under magnetic stirring until pH 7was reached.. By varying the chitosan concentration in solution (0.1%, 0.2% and 0.5%, w/v), three batches of microparticles with different chitosan contents were prepared (9.1%, 16.7%, and 33.3%, w/w). Separation of chitosan-coated microparticles from aqueous phase was done by centrifugation at 1000 rpm for 5 min (302 K, Sigma, Germany). After removing the supernatant, the product was washed with ultra-pure water and centrifuged again. This washing and centrifugation step was repeated three times. The product was dried in a vacuum oven and sieved to yield 2 size fractions (<90 µm, and 125-250 µm).

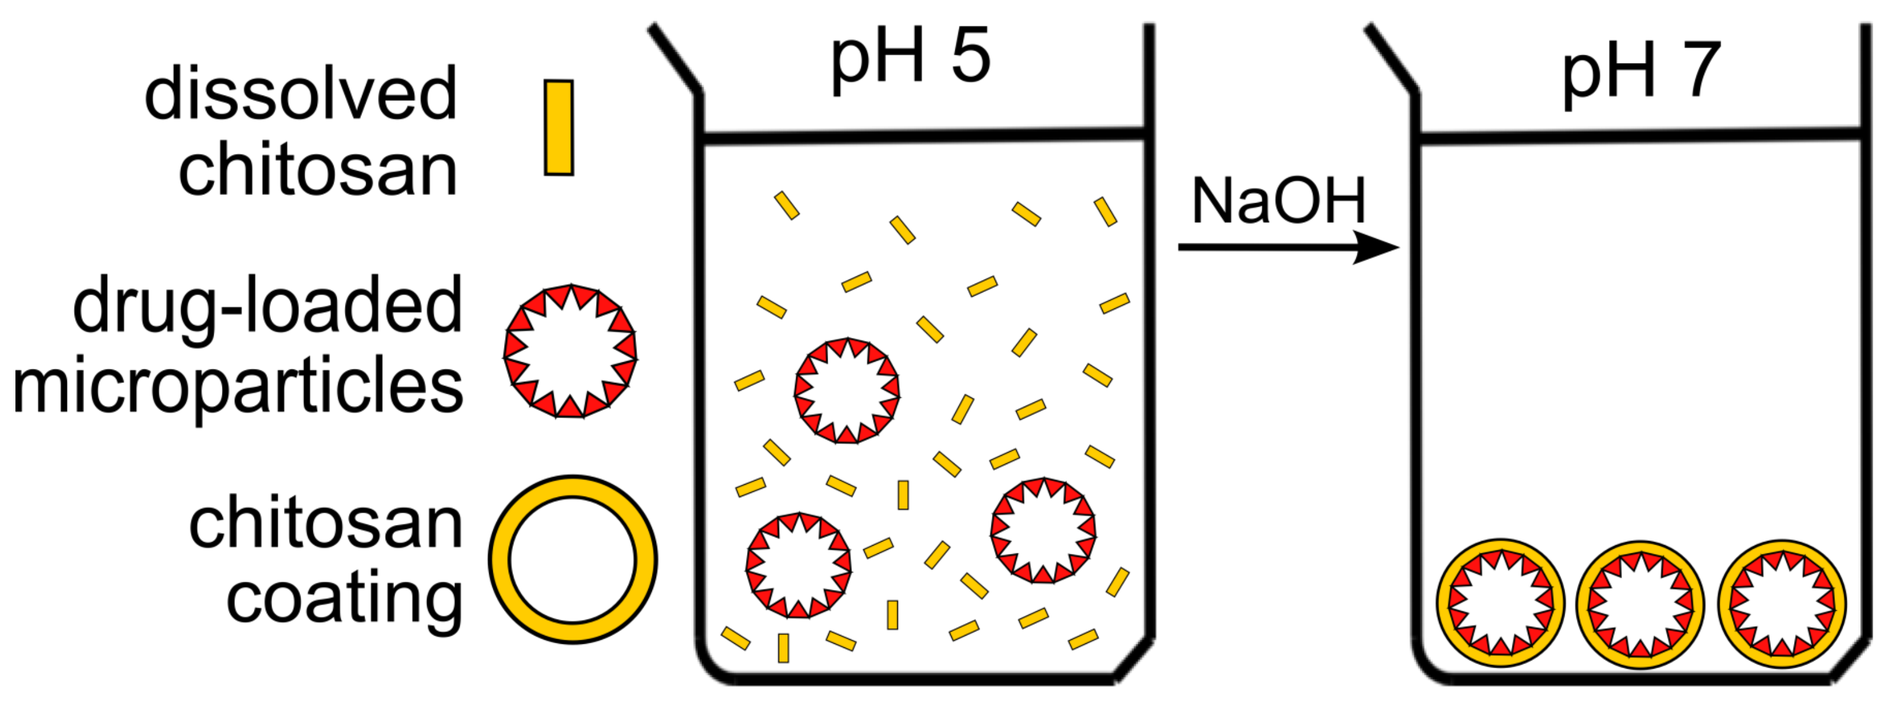
dissolved chitosan
drug-loaded microparticles
chitosan coating
pH 5
NaOH
pH 7

Fig. 2: Schematic representation of the chitosan precipitation method.

Particle characterization

Coating quality was visually analyzed by scanning electron microscopy (SEM). Drug-load quantification was done by a validated HPLC method [6]. FCC content of chitosan-coated microparticles was determined by calcium quantification using capillary electrophoresis. Chitosan quantification was performed by a colorimetric assay described by Larionova *et al*. [10]. Drug release was measured using the USP IV flow-through method in phosphate buffer (pH 6.8). Sample mass was 50 mg, and the flow rate was set to 16 mL/min.

Particle-retention assay

The flow channel for measuring particle retention was adapted from Batchelor *et al*. [9], and designed and built in-house (**Fig. 3**). It consisted of three main parts, i.e. a) upper plate, b) fixation plate, and c) support plate. Porcine colonic mucosa was spread on the mucosa holder (d) of the support plate and immobilized with the fixation plate. After a pre-hydration phase of 5 min with a constant flow of ultra-pure water (37°C), 20.0 mg of particles was evenly distributed over the center area of the mucosa (see **Fig. 4**). The flow channel was kept in horizontal position for 5 min. Subsequently, the assembly was tilted to 45° and the flow was started. The outcoming flow medium was collected in beakers which were changed every 10 min. The duration of the experiment was set to 30 min. The remaining particles were scraped-off the mucus for determination of FCC recovery.

Particle-retention assay

Particle retention quantification was done by analysis of the carrier particle (FCC). The collected flow medium was adjusted to pH 2.5 with HCL leading to chemical dissolution of calcium carbonate. The calcium ions in solution were then quantified by capillary electrophoresis, and the mass of FCC was determined by means of a calibration curve. The percentage of detached FCC (FCC_{det}) was calculated by Eq. 1, where FCC_{app} is the amount of FCC initially applied on the mucosa. The relative adhesion to mucosal tissue is presented as retained FCC (FCC_{ret}) according to Eq. 2.

$$FCC_{det}(\%) = \frac{FCC_{det}(mg)}{FCC_{app}(mg)} x100 \quad (1)$$

$$FCC_{ret}(\%) = 100\% - FCC_{det}(\%) \quad (2)$$

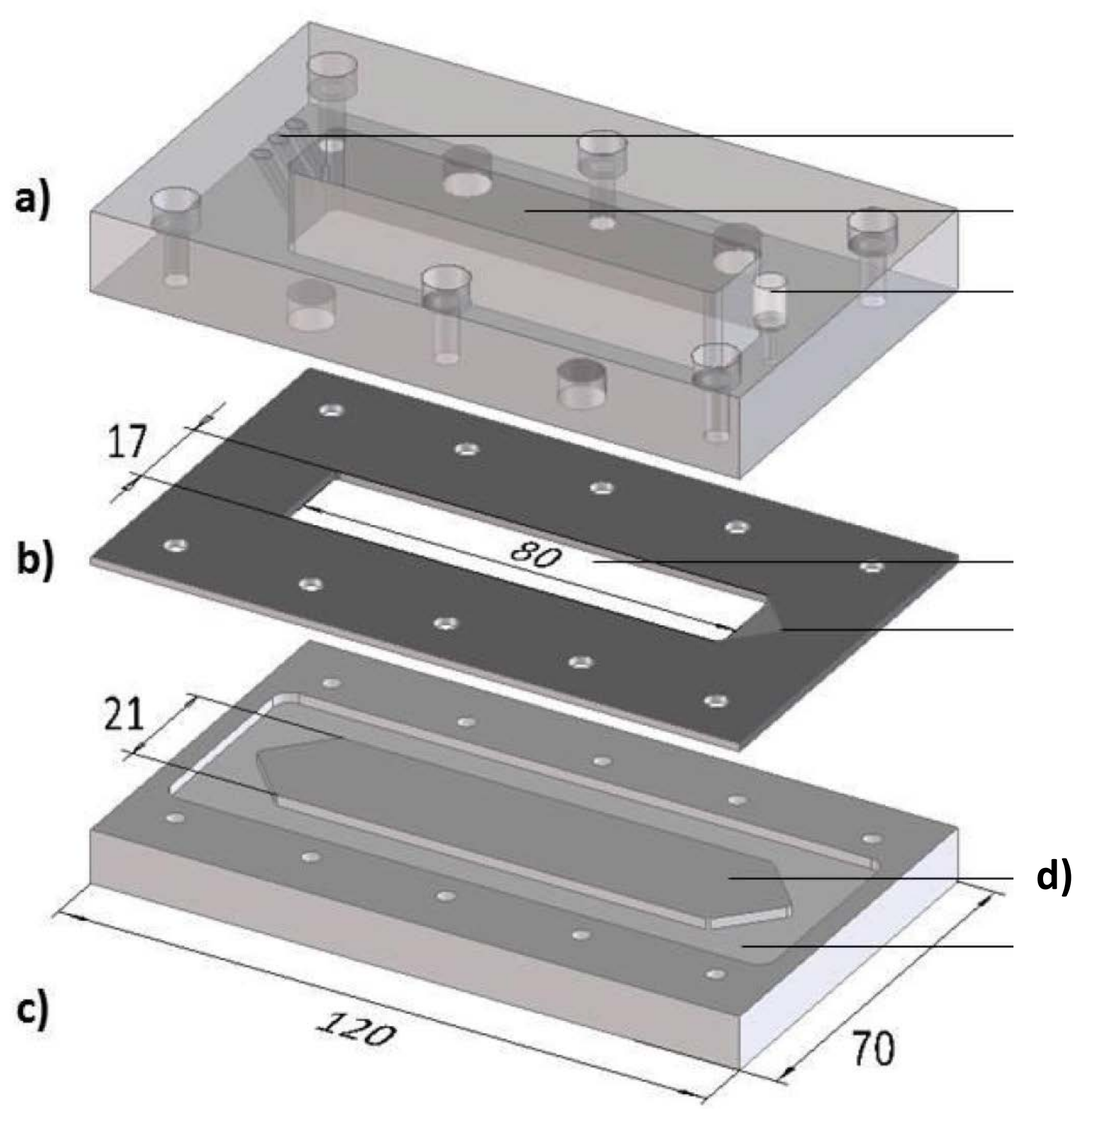
a)
b)
c)
d)
17
80
21
120
70

Fig. 3: Flow channel assembly with dimensions in mm.

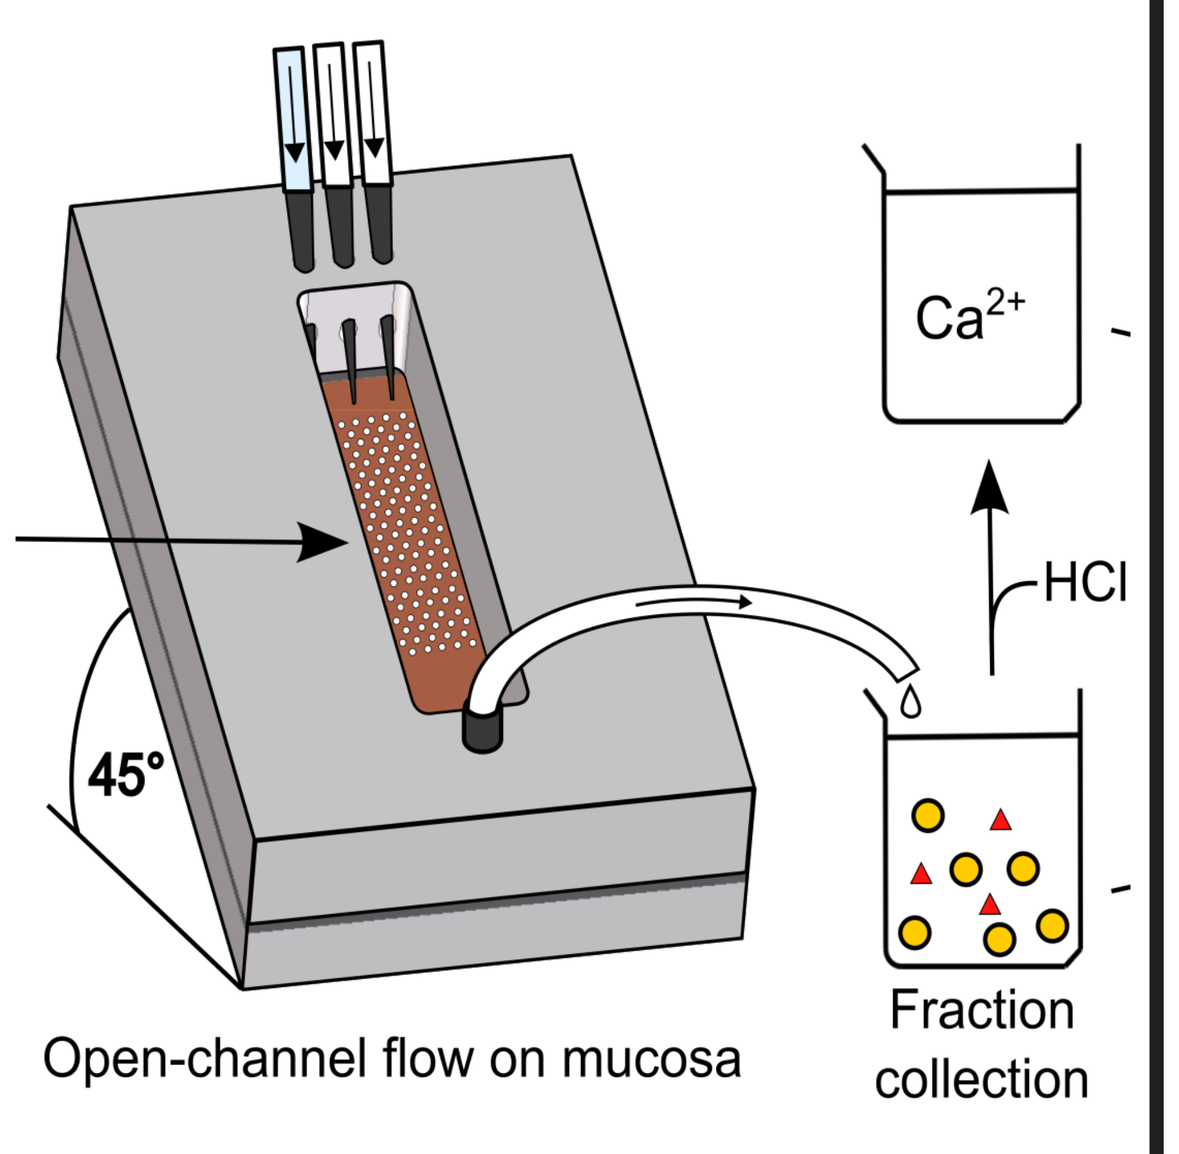
Ca2+
HCl
45°
Open-channel flow on mucosa
Fraction collection

Fig. 4: Particle-retention assay schematic representation.

RESULTS & DISCUSSIONS

Particle characterization

SEM images of non-mucoadhesive control particles (MBZ-loaded FCC) are shown in **Fig. 5** (A: < 90 µm, B: 125 – 250 µm). Fig. 5 also shows MBZ-loaded FCC coated with 33% (w/w) chitosan (C: < 90 µm, D: 125 – 250 µm). The distinct pore structure of FCC was completely covered indicating the formation of micrometer-thick chitosan layers. The particle compositions after chitosan precipitation were consistent with expected values demonstrating a good control of the deposition process (see **Fig. 6**).

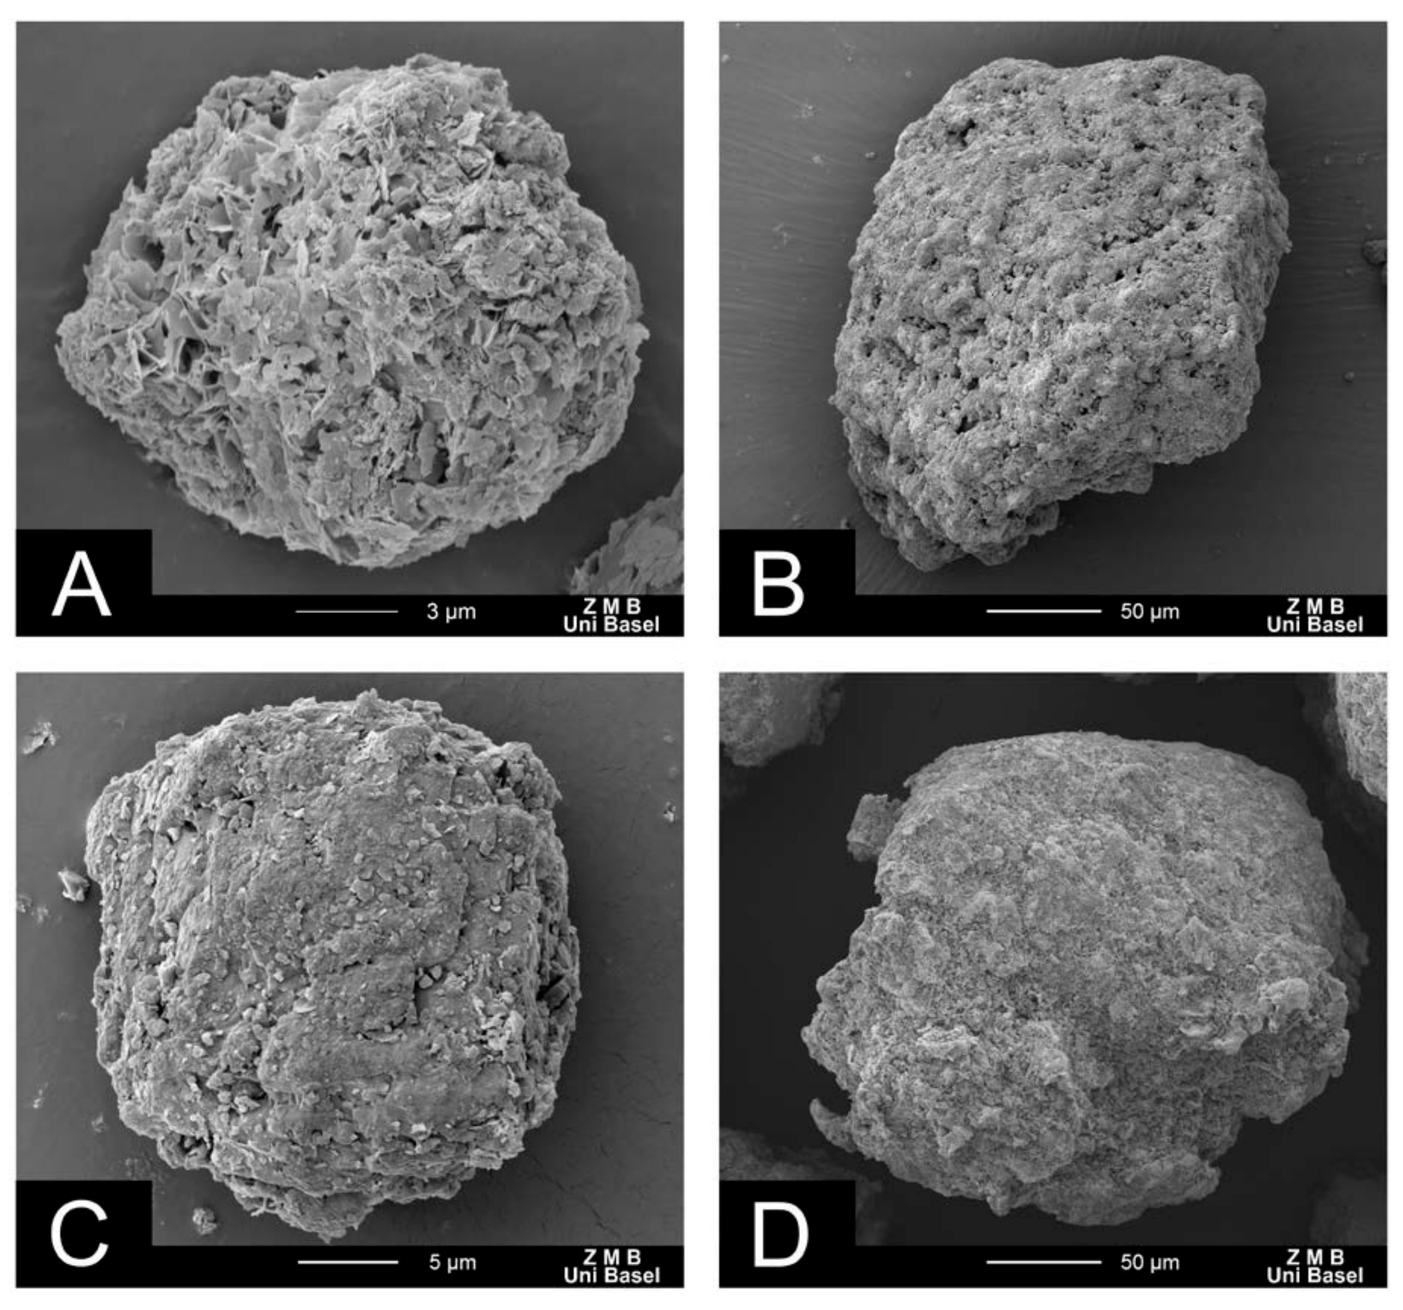
A
B
C
D

Fig. 5: SEM images of drug-loaded FCC (A&B) and chitosan-coated FCC (C&D).

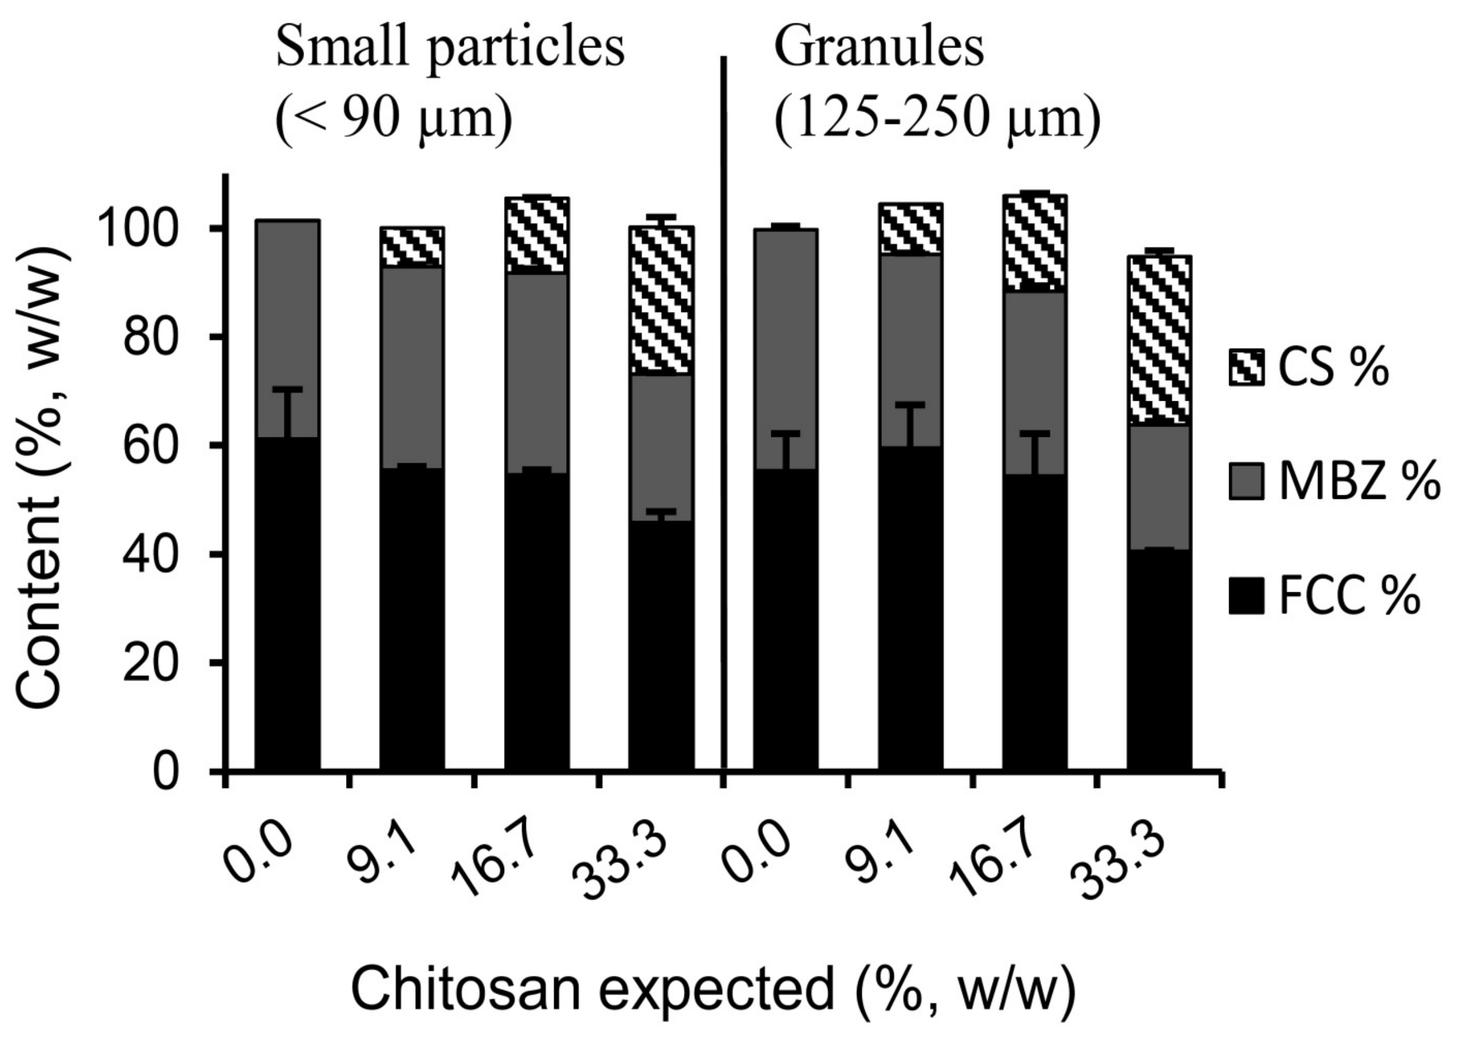
Small particles (< 90 µm)
Granules (125-250 µm)
Content (%, w/w)
CS %
MBZ %
FCC %
0.0
9.1
16.7
33.3
Chitosan expected (%, w/w)

Fig. 6: Particle composition (n=3).

Particle-retention assay

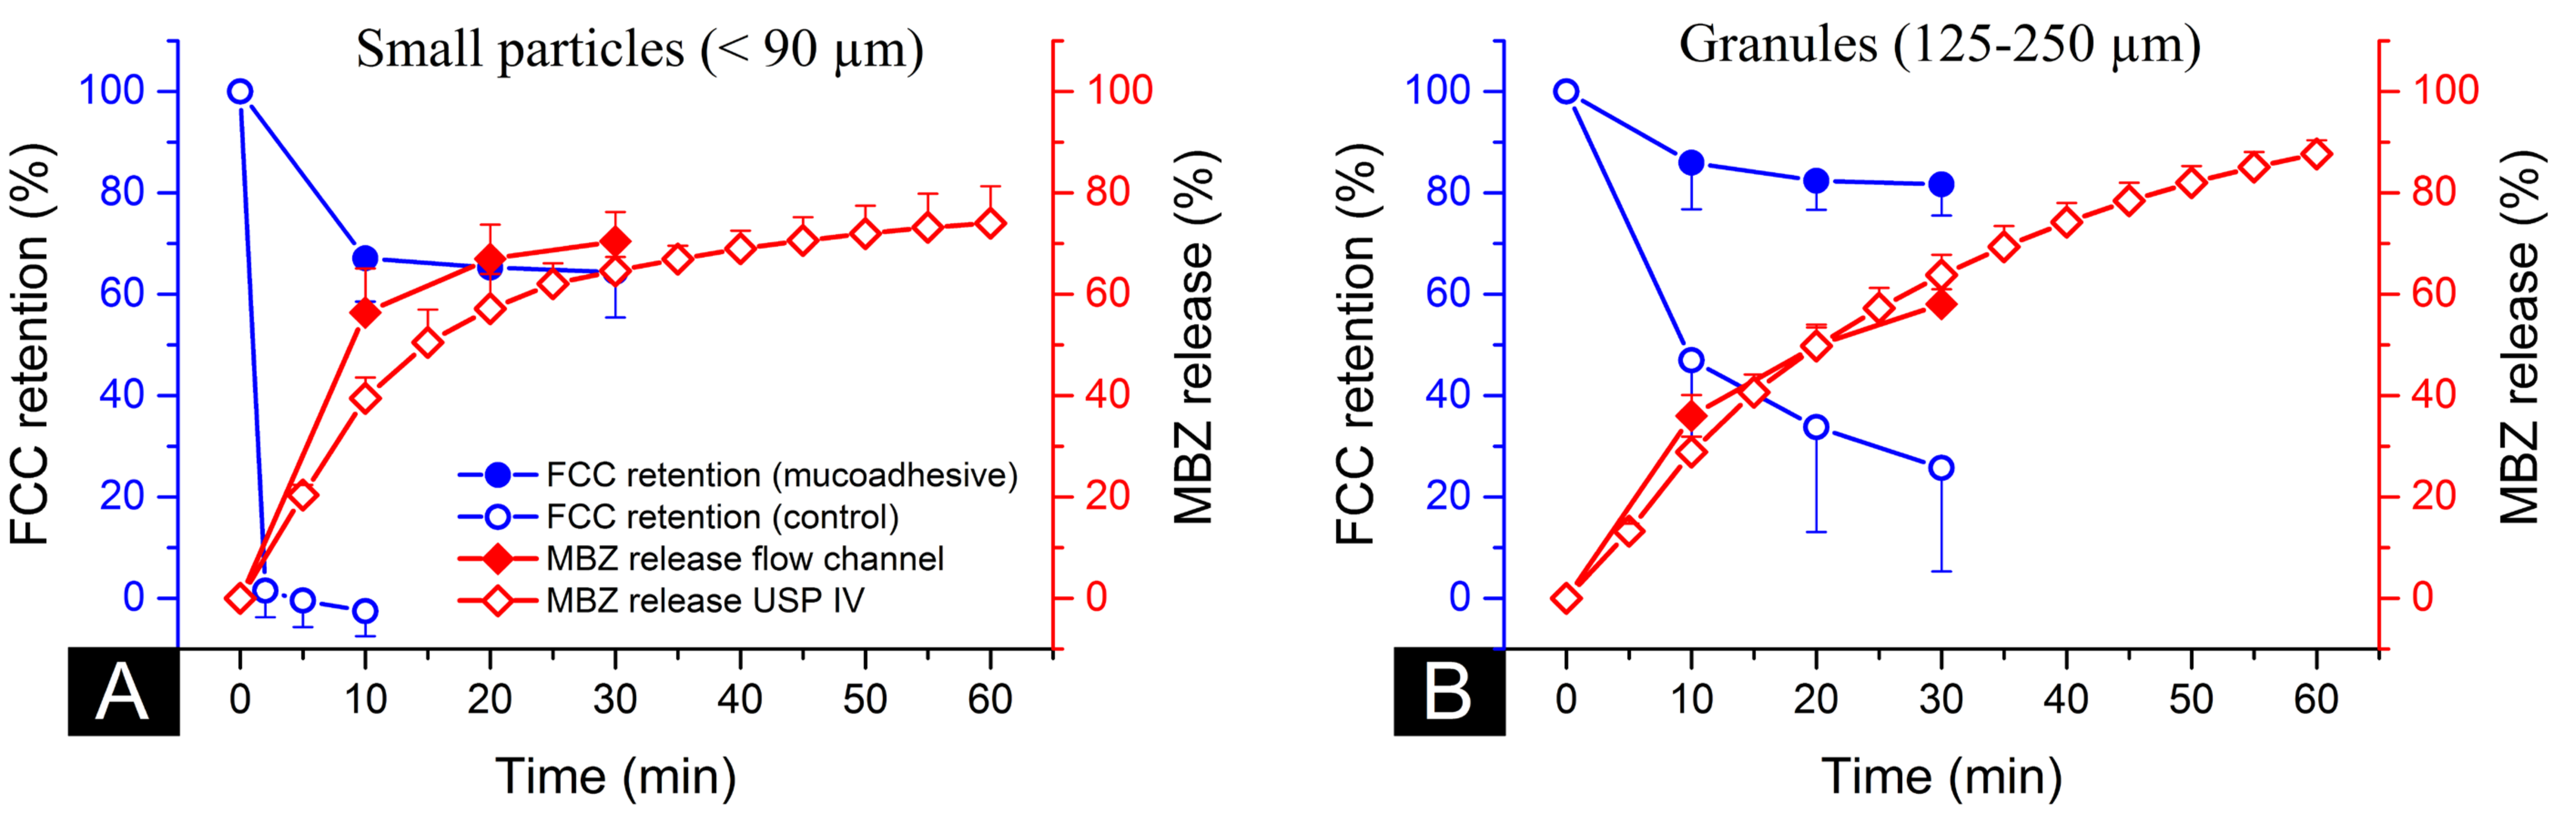
Small particles (< 90 µm)
Granules (125-250 µm)
FCC retention (%)
MBZ release (%)
FCC retention (mucoadhesive)
FCC retention (control)
MBZ release flow channel
MBZ release USP IV
A
B
Time (min)

Fig. 7: Results particle-retention assay for 33.3% chitosan (n=5). A) small particles (< 90 um) B) granules (125-250 um).

Particle-retention assay

The results in **Table 1** show that mucoadhesion strongly depended on the extent of chitosan coating. Mucoadhesive granules (125-250 µm) with a chitosan content of 33.3% (w/w) showed strongest retention on colonic mucosa (81.6 ± 6.2%, w/w). Immediate washout of the control particles (< 90 µm) was observed after only 2 min. Most values for FCC recoveries were between 84.6% and 96.5% (w/w), indicating a good reliability of the FCC quantification method.

Table 1: Particle retention after 30 min ($FCC_{ret,30}$) of mucoadhesive particles with different chitosan contents (n=5).

	Small particles (< 90 µm)		Granules (125-250 µm	
Chitosan (%, w/w)	$FCC_{ret,30}$ (%, w/w)	Recovery (%, w/w)	$FCC_{ret,30}$ (%, w/w)	Recovery (%, w/w)
0	-2.6 ± 4.9	115.5 ± 6.5	25.7 ± 20.4	105.2 ± 11.1
9.1	27.6 ± 7.0	96.0 ± 5.8	62.0 ± 14.9	89.7 ± 18.0
16.7	32.3 ± 7.4	92.1 ± 11.7	69.2 ± 6.5	96.5 ± 6.8
33.3	64.4 ± 9.0	67.7 ± 7.6	81.6 ± 6.2	84.6 ± 23.1

Particle-retention and drug-dissolution kinetics for the particles with an expected chitosan content of 33.3% (w/w) are shown in **Fig. 7**. The retention kinetics of mucoadhesive particles were characterized by an initial burst detachment within the first 10 min, followed by lower detachment rate for the remaining time of the experiment. The two different dissolution methods (flow-channel and USP IV) showed comparable drug-release kinetics for tested particles.

REFERENCES

[1] Varum *et al.*, Int. J. Pharm., 2011.
[2] Efentakis *et al.*, AAPS PharmSciTech, 2000.
[3] Varum *et al.*, Crit. Rev. Ther. Drug Carrier Syst., 2008.
[4] He *et al.*, Int. J. Pharm., 1999.
[5] Ali *et al.*, J. Adv. Pharm. Technol. Res. 5, 2014.
[6] Preisig *et al.*, Eur. J. Pharm. Biopharm., 2014.
[7] Khutoryanskiy, Macromol. Biosci., 2011.
[8] Han *et al.*, Macromol. Rapid Commun., 2012.
[9] Batchelor *et al.*, Int. J. Pharm., 2002.
[10] Larionova *et al.*, Carbohydr. Polym., 2009.